Finafloxacin demonstrates utility in treating *Burkholderia pseudomallei in vivo*

Kay B. Barnes[1], Mark I. Richards[1], Andreas Vente[2] and Sarah V. Harding[1]

Defence Science and Technology Laboratory, Porton Down, UK[1]
MerLion Pharmaceuticals, Berlin, Germany[2]

Introduction

B. pseudomallei is intrinsically resistant to many antimicrobials, most apparent in acidic conditions that may be encountered *in vivo.* Finafloxacin has been chemically altered to improve its activity under acidic conditions, where other fluoroquinolones are less active. Finafloxacin has demonstrated a significant benefit when compared to co-trimoxazole d in a mouse model of melioidosis (see poster presented by Kay Barnes). Further analysis has been performed on tissues harvested from this study.

Methods

1. Balb/c mice were infected with 98 CFU of *B. pseudomallei* by the inhalational route and antibiotics initiated at 24 and 36 hours post-challenge (hpc).
2. Five mice per group were sacrificed following 1 or 14 days of treatment and organs harvested for analysis.
3. Tissues fixed in buffered formalin were embedded in paraffin wax and 4 µm sections stained with H &E. A semiquantitative assessment of lesions was performed where; 1 = Minimal pathology, 2 = Mild, 3 = Moderate, 4 = Severe.
4. The levels of TNFα, IL-6, IL-1β, 1L-10 and IL-12 were determined on 50 µL of plasma using the Bio-plex Pro Mouse Cytokine 23-plex assay.

Results

When 1 day of treatment was initiated at 24 hpc, the spleen, liver and lungs harvested from mice treated with finafloxacin and the lungs from mice treated with co-trimoxazole weighed less than the controls (Figure 1A + B). The lungs from mice treated with finafloxacin were lighter than those treated with co-trimoxazole.

When 14 days of treatment was initiated at 24 hpc, the spleen and lungs harvested from mice treated with finafloxacin weighed less than those treated with co-trimoxazole (Figure 1C + D). The same differences were observed in the spleen and lungs when treatment was initiated at 36 hpc.

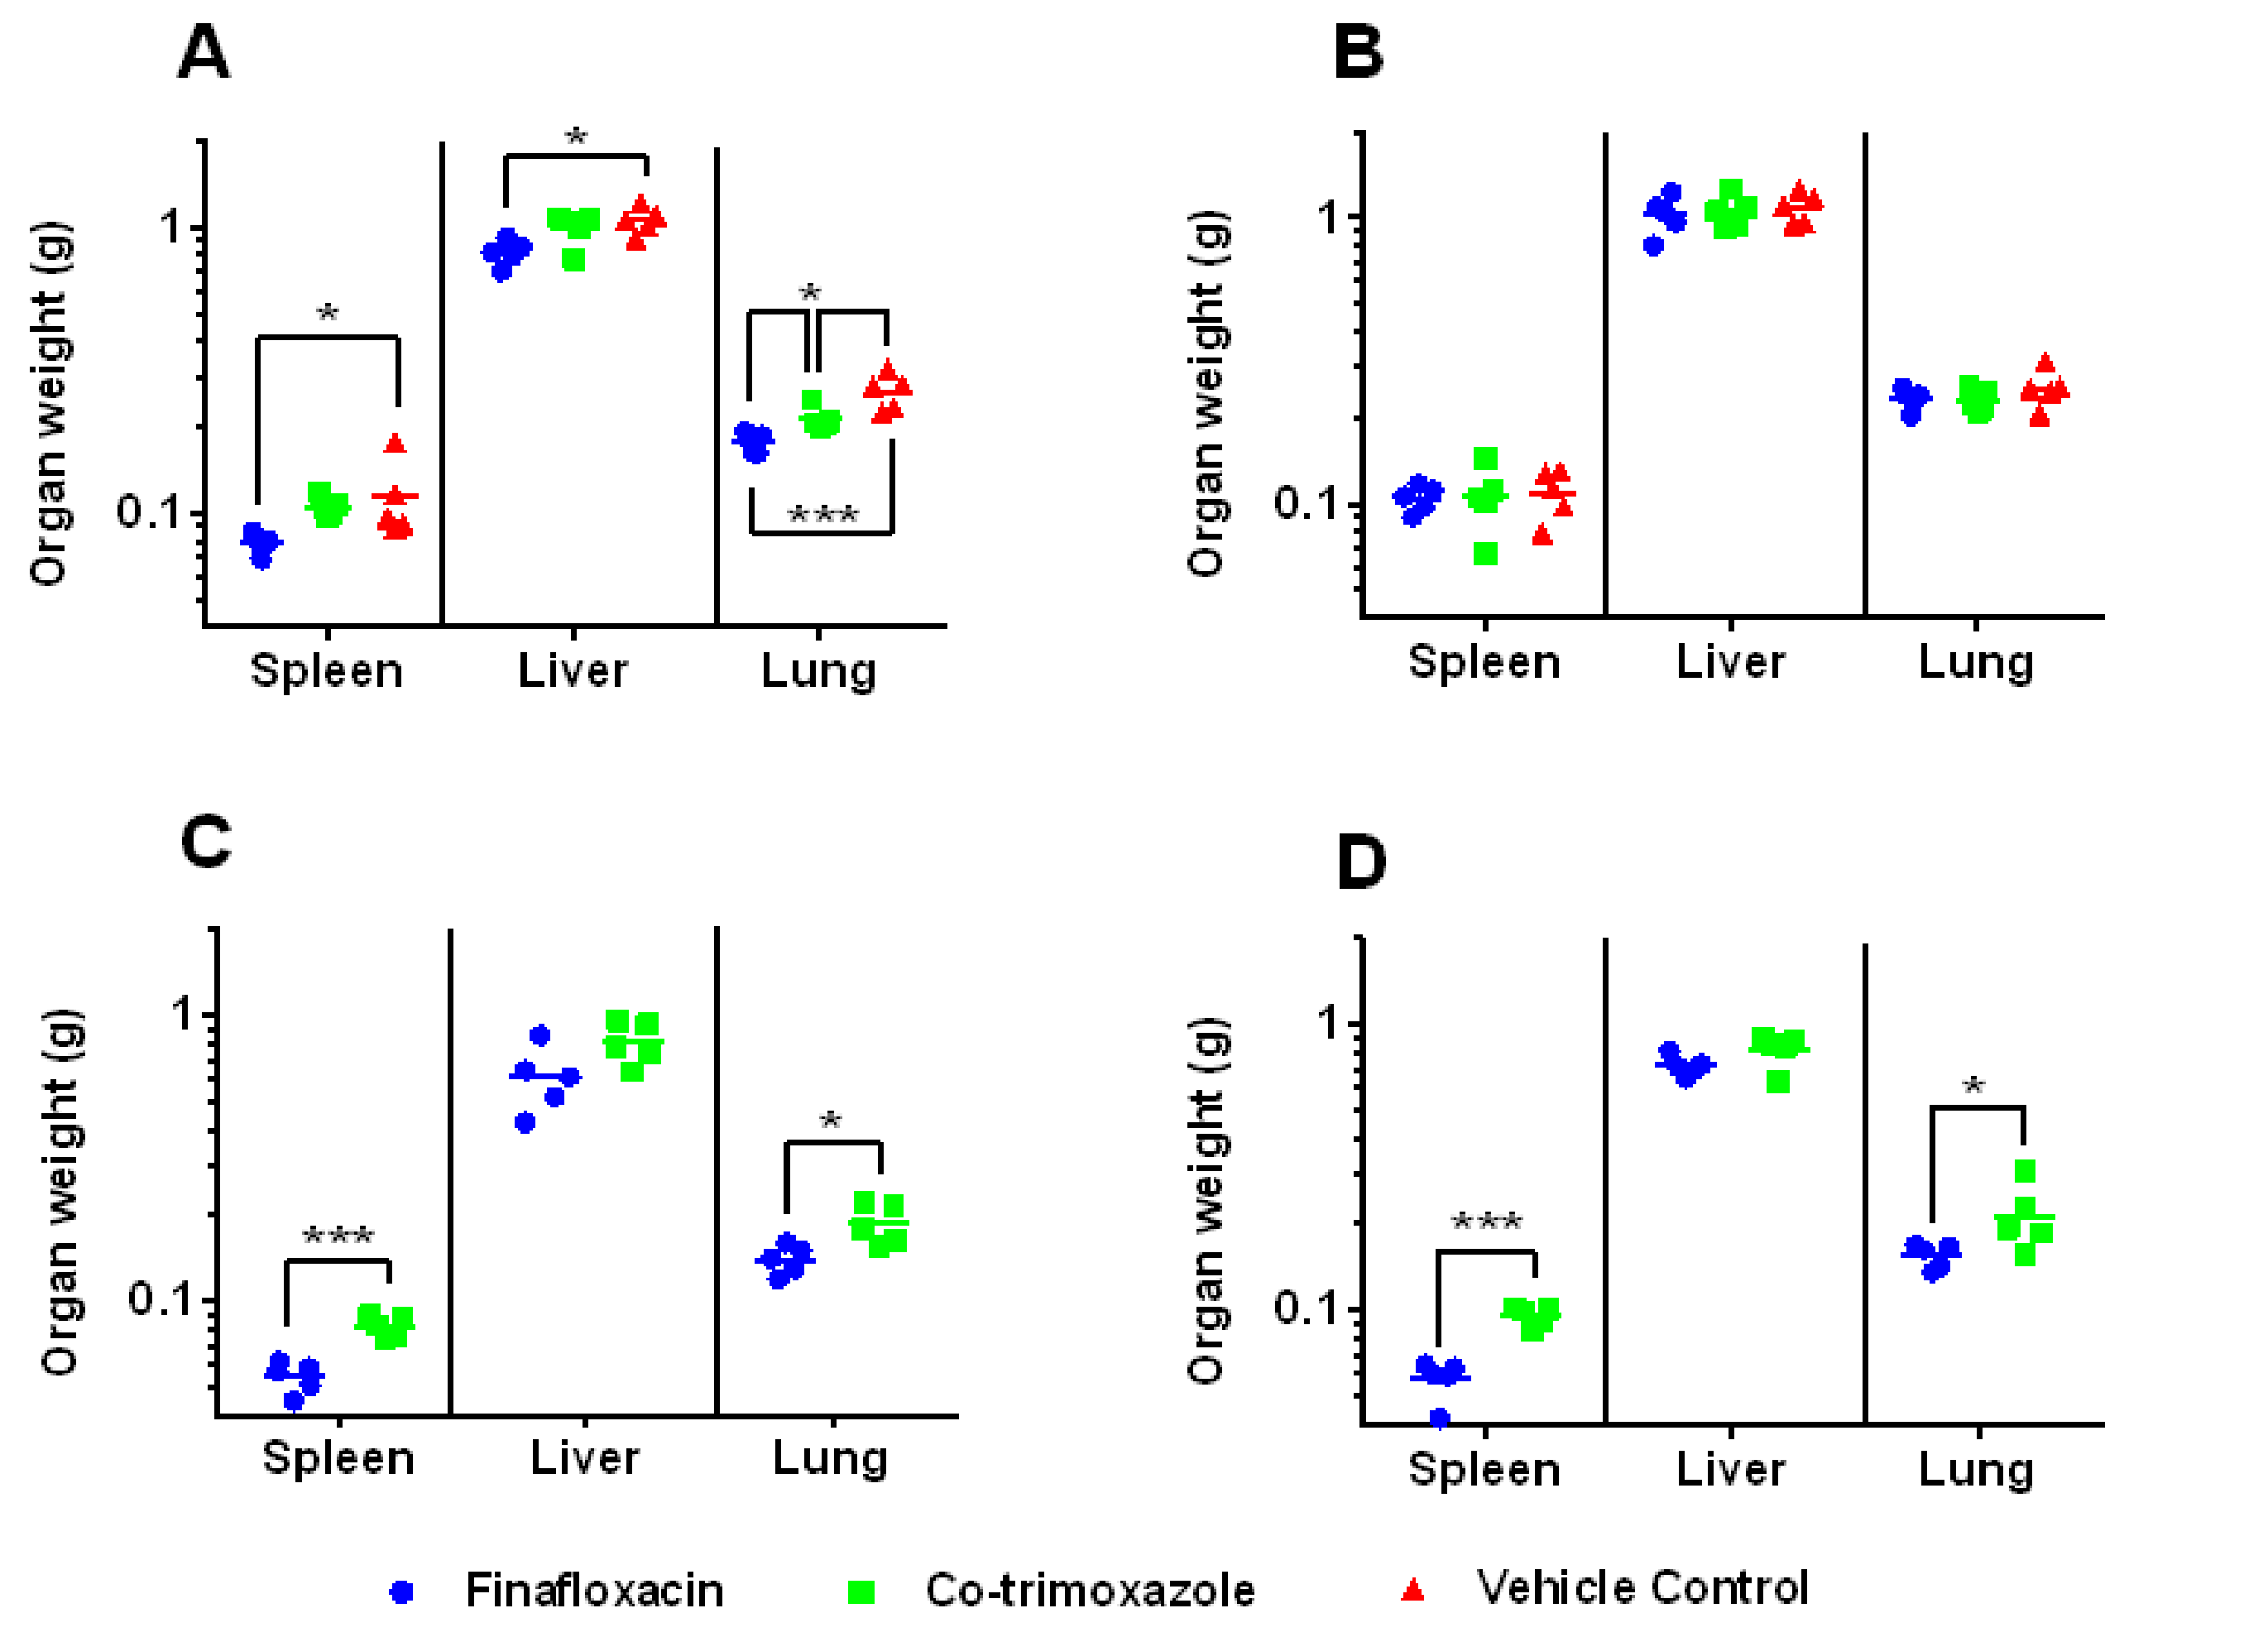

A
B
C
D
Organ weight (g)
Spleen
Liver
Lung
Finafloxacin
Co-trimoxazole
Vehicle Control

Figure 1. The organs weights following antibiotic treatment. The weight (g) of the organs from 5 mice per group following one day of treatment initiated at 24 (A) or 36 (B) hpc or 14 days of treatment initiated at 24 (C) or 36 (D) hpc. *p<0.05, ***p<0.001.

None of the antibiotic treated animals had lesions in the spleen, compared to the controls (Figure 2). The frequency and severity of acute lesions in the liver were significantly reduced in the antibiotic treated animals when compared with the controls. Finafloxacin was more effective in preventing the onset of hepatic lesions in mice treated at 24 hpc and culled following one day of antibiotic treatment, this trend was reversed in mice treated at 36 hpc where co-trimoxazole significantly reduced liver pathology (Figure 2). All animals treated at 24 or 36 hpc and culled following 14 days of treatment. had some degree of residual lesions with similar severity.

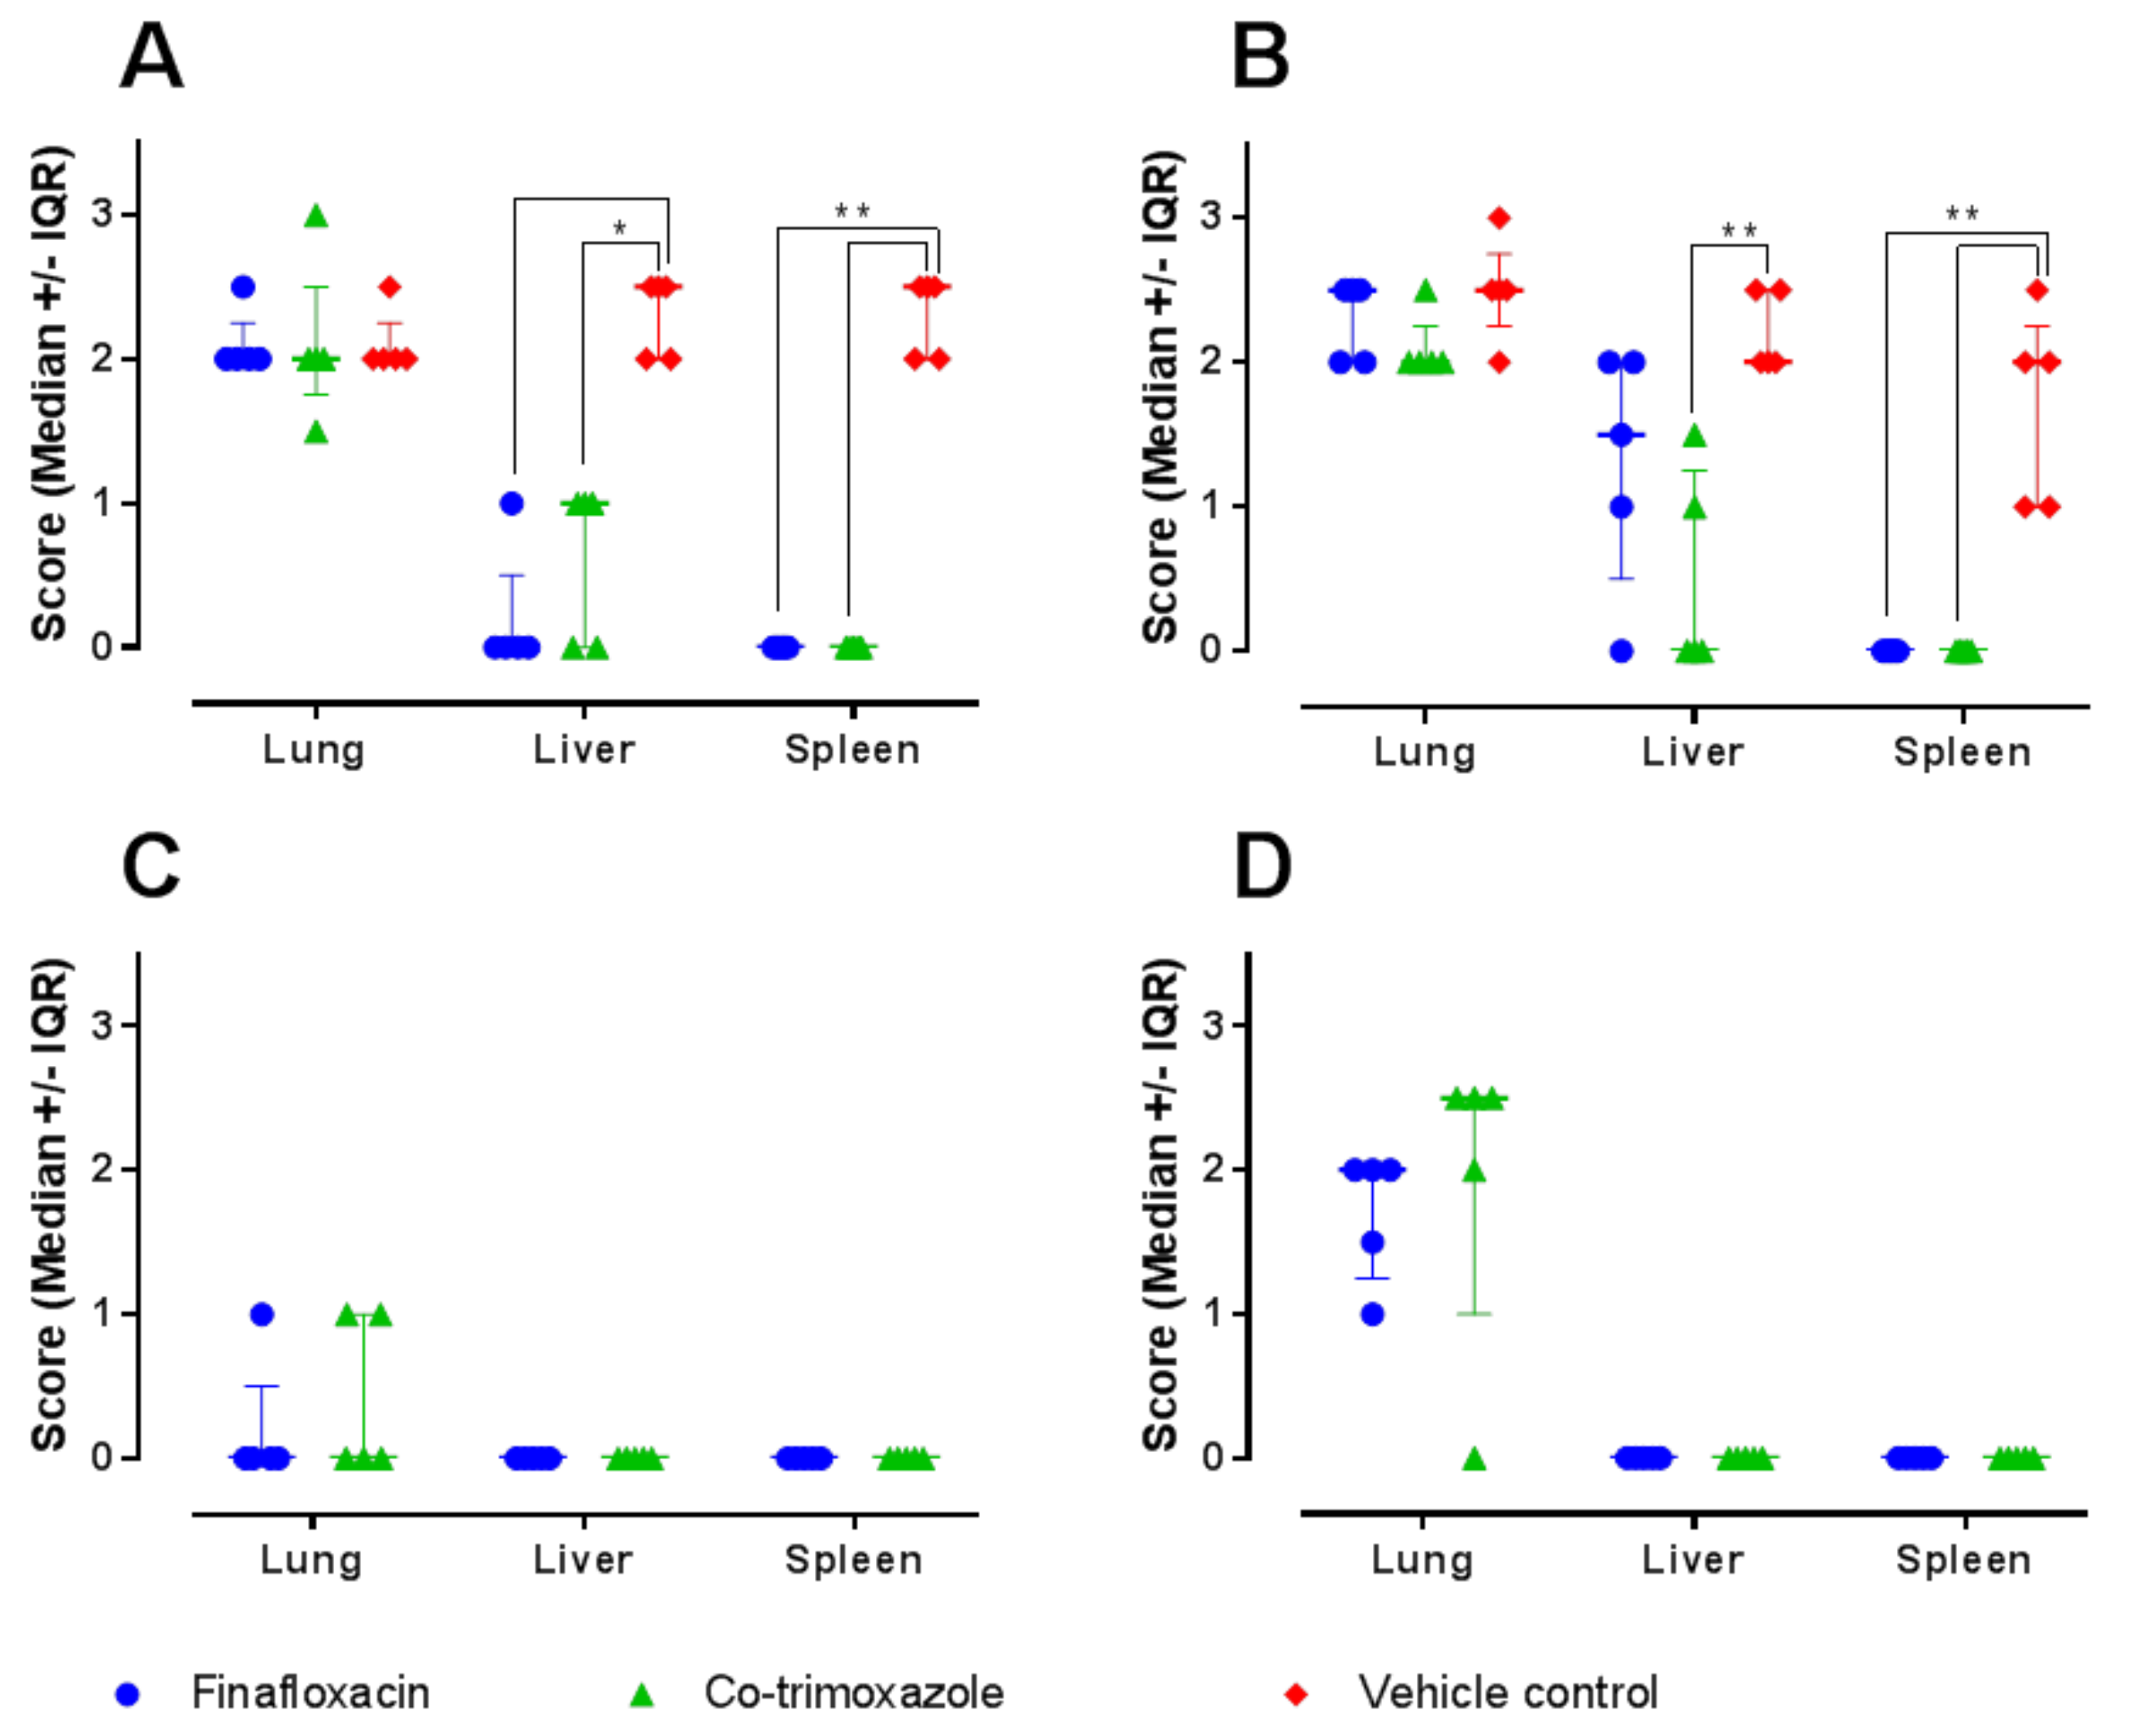

A
B
C
D
Score (Median +/- IQR)
Lung
Liver
Spleen
Finafloxacin
Co-trimoxazole
Vehicle control

Figure 2. The histopathological scores of organs following antibiotic treatment. Scores were given to the appearance and structure of the organs from 5 mice per group following one day of treatment initiated at 24 (A) or 36 (B) hpc or 14 days of treatment initiated at 24 (C) or 36 (D) hpc. *p<0.05, **p<0.01.

Some animals treated at 24 hpc had minimal residual lesions in the lungs compared to those treated at 36 hpc that had significant pathology (Figure 2). Animals treated at 36 hpc for 14 days presented with chronic lesions in the lung. Multinucleated giant cells and encapsulated granulomas around a central core of necrosis were not observed.

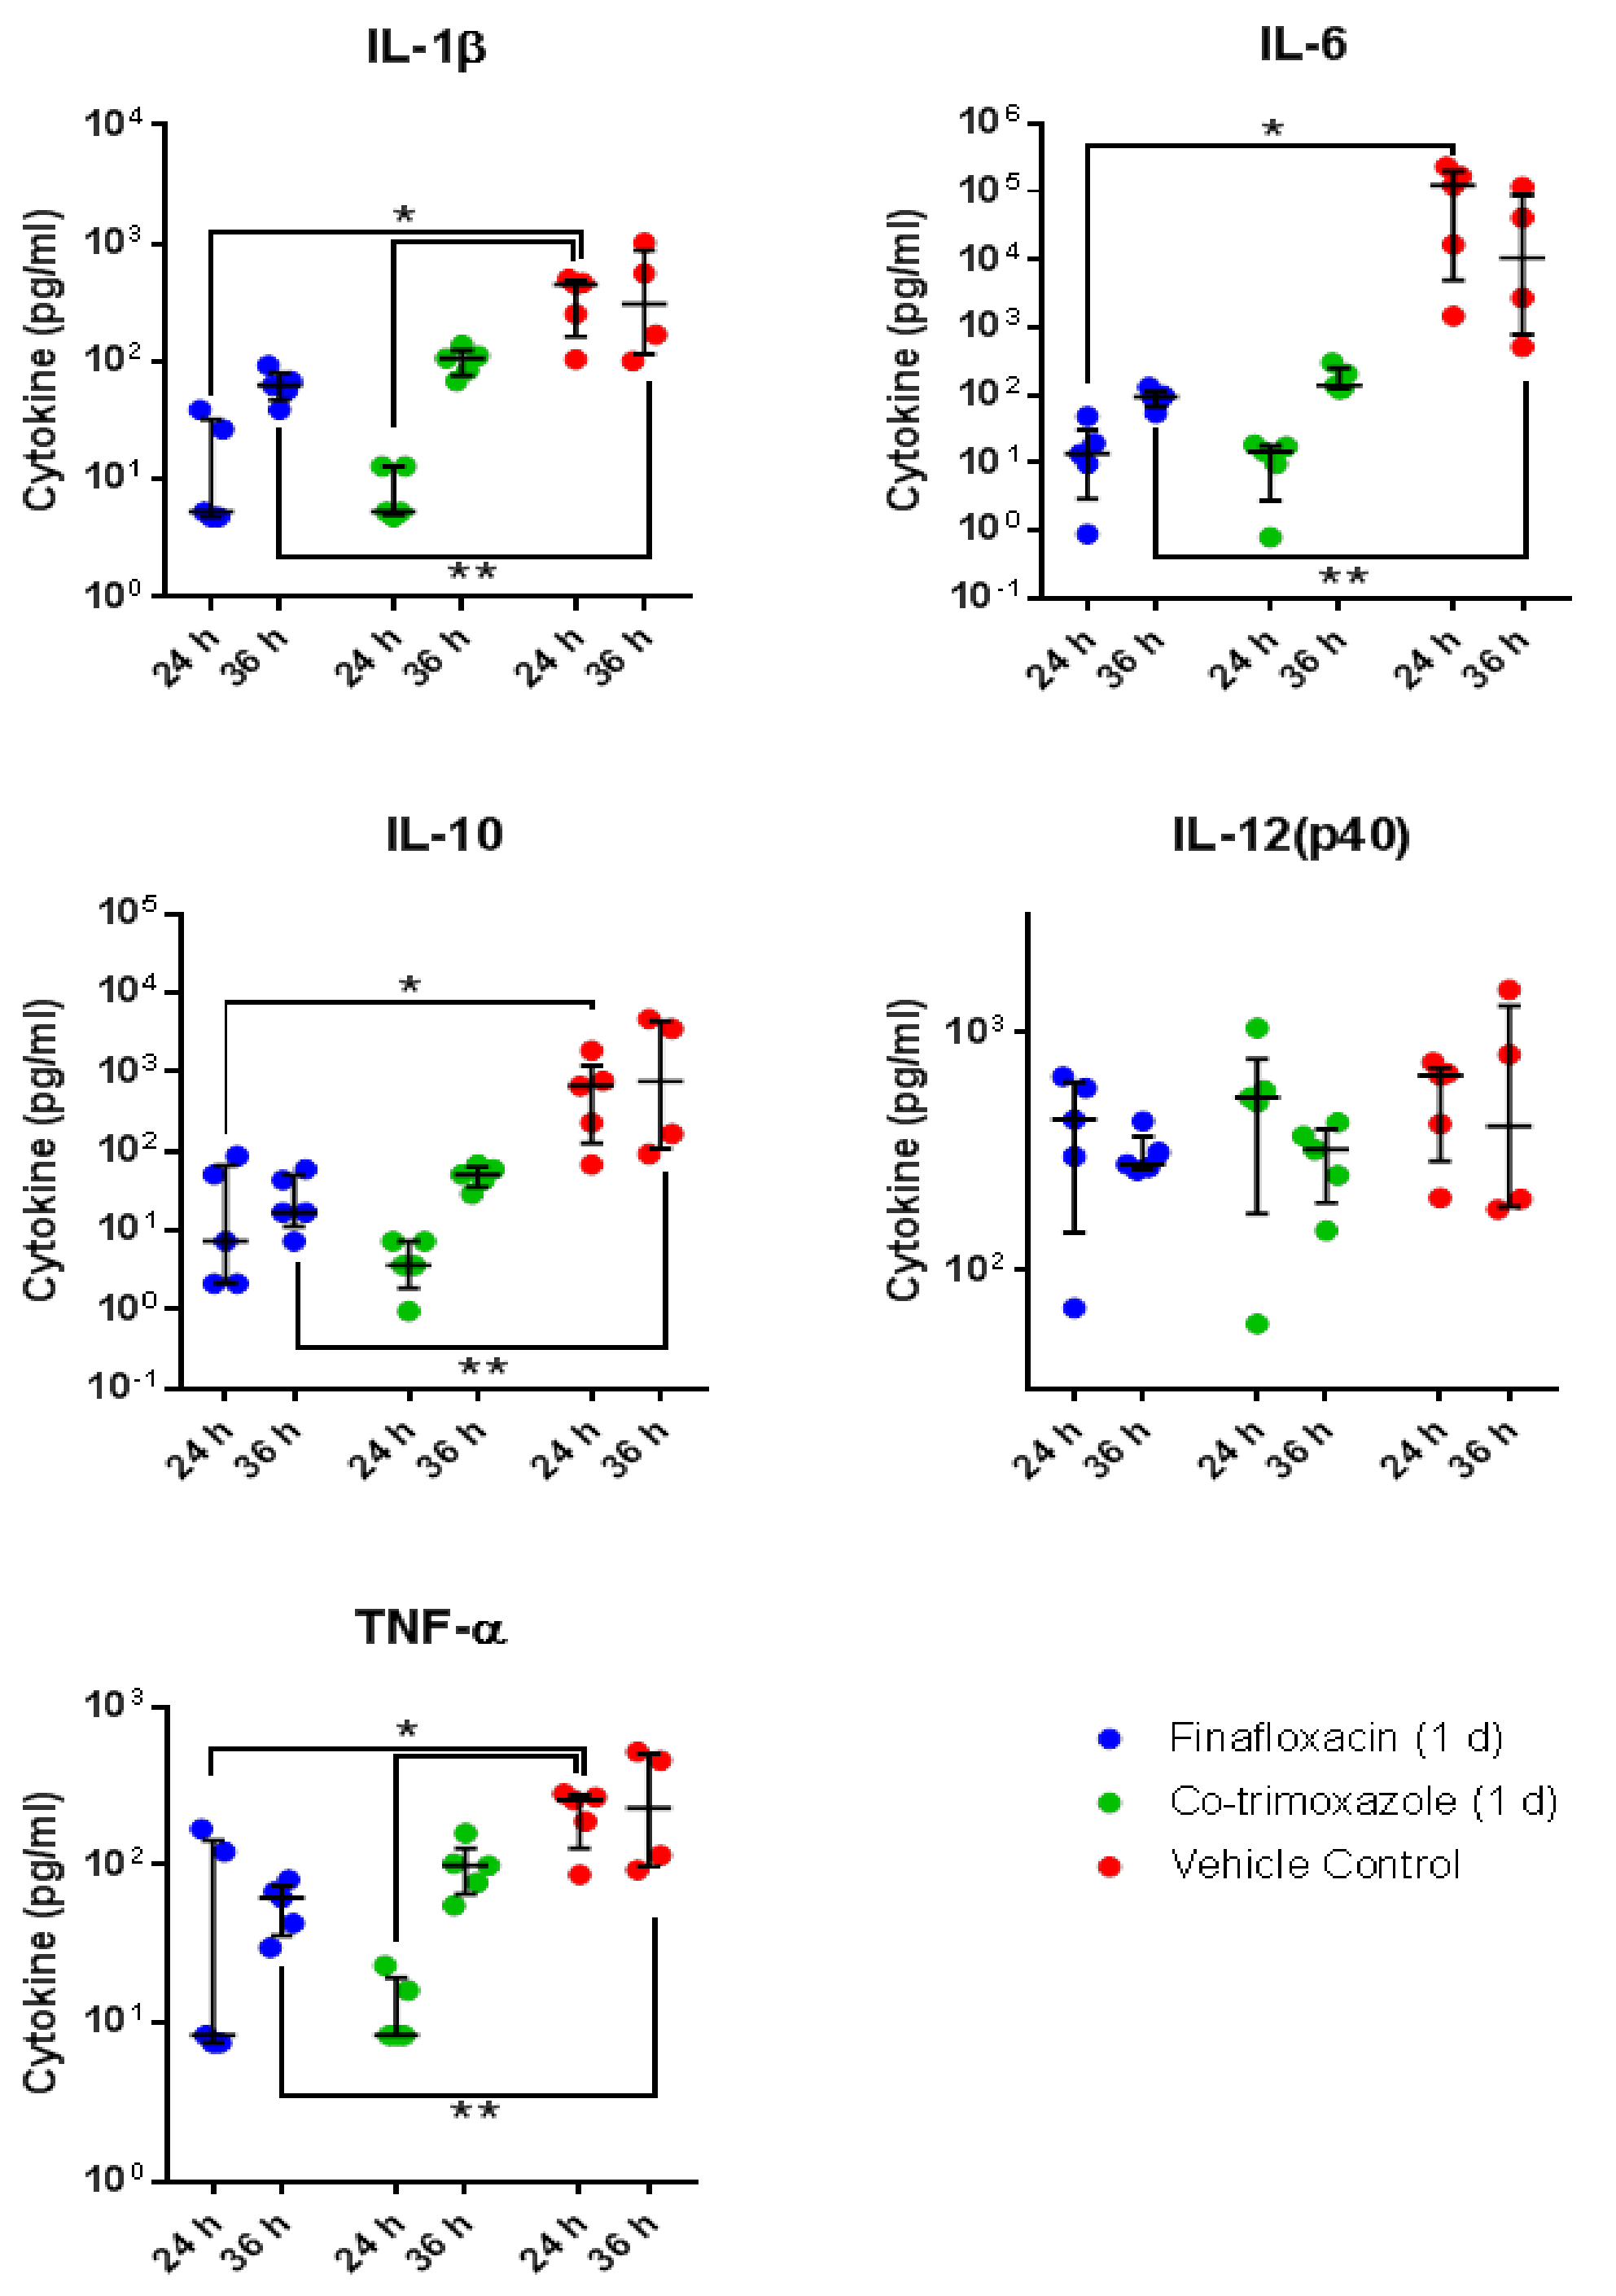

IL-1β
IL-6
IL-10
IL-12(p40)
TNF-α
Cytokine (pg/ml)
24 h
36 h
Finafloxacin (1 d)
Co-trimoxazole (1 d)
Vehicle Control

Figure 3. The cytokine levels in the plasma following one day of antibiotic treatment. Cytokine levels were determined in the plasma of 5 mice per group following one day of antibiotic treatment initiated at 24 or 36 hours post-challenge.*p<0.05, **p<0.01.

The levels of IL-6, IL-1β, 1L-10 and TNFα were significantly reduced in mice receiving one day of finafloxacin treatment initiated at 24 or 36 hpc when compared to the controls (Figure 3). The levels of IL-1β and TNFα were significantly reduced in mice receiving co-trimoxazole at 24 hpc. No differences were observed in the level of IL-12. In addition, no differences were observed between the antibiotic treated groups.

Discussion

Finafloxacin has demonstrated a significant protective benefit when compared to co-trimoxazole in a mouse model of melioidosis (see poster presented by Kay Barnes). The additional data generated to date has identified differences between the antibiotic treated animals and the controls but also differences between treatments. Further investigation is required to determine the location of *B. pseudomallei* in antibiotic treated animals following cessation of therapy (at day 14) and before relapse of disease (day 50).

This work was funded by DTRA under contract HDTRA1-12-D-0003 0019.

© Crown copyright (2018), Dstl. This material is licensed under the terms of the Open Government Licence except where otherwise stated. To view this licence, visit http://www.nationalarchives.gov.uk/doc/open-government-licence/version/3 or write to the Information Policy Team, The National Archives, Kew, London TW9 4DU, or email: psi@nationalarchives.gsi.gov.uk.

DSTL/PS106886