A1-685

Determination of the Effect of Age and Gender on the Pharmacokinetics (PK) and Tolerability of a Single Dose of Finafloxacin HCl (FIN) in Healthy Volunteers

L. Mooney[1], M. Murphy[2], M. Kabbaj[3], A. Vente[1], H. Labischinski[1], and D. Swearingen[4]

[1]MerLion Pharmaceuticals GmbH, Berlin, Germany, [2]Celerion, Belfast, United Kingdom, [3]Celerion, Zurich, Switzerland, [4]Celerion, Tempe, AZ.

Contact:
Andreas Vente
MerLion Pharmaceuticals GmbH
Robert-Roessle-Str. 10
D-13125 Berlin
Germany
T.+49-30-9489-4050
vente@merlionpharma.de

Abstract

Introduction: FIN is a novel fluoroquinolone that exhibits improved antibacterial and pharmacodynamic properties under acidic conditions which often characterize infection sites. Previous clinical studies have indicated that FIN is well tolerated with few treatment related adverse events (AEs). This study aimed to determine primarily the effect of age and gender on the PK of FIN and assess its safety profile.

Methods: The study was conducted under approval of the Celerion IRB, the FDA IND # 106,696, and in accordance with GCP. This was a single center, open label study in 40 healthy subjects (10 young adult males, 10 young adult females, 10 elderly adult males, and 10 elderly adult females). All subjects received a single oral dose of 400 mg FIN (2 x 200 mg tablets).

Results: Following a single dose of 400 mg, the mean peak exposure (C_{max}) of FIN was similar in elderly (4630 - 6190 ng/mL) and young (5160 - 6440) subjects. The average exposure (AUC_{0-inf}) was approximately 18% greater in elderly (20250 - 23477 ng.h/mL) versus young (17240 - 19942 ng.h/mL) subjects (not statistically significant). Mean exposure was similar in males and females (<16% difference) and Cmax was statistically higher in females (6190 - 6440 ng/mL) than males (4630 - 5160 ng/mL). The mean $t_{1/2}$ of FIN was comparable in both genders within each age group. Renal clearance was reduced in the elderly group. In total, 23 AEs (22 mild and 1 moderate) and no serious AEs were observed. There were no significant findings in clinical laboratory values, vital signs, ECGs, and physical examinations.

Conclusions: There were no statistically significant age or gender effects on FIN PK except on urinary excretion (age) and peak exposure (gender). The administration of a single oral dose of 400 mg FIN was safe and well tolerated in the young and elderly male and female subjects in this study.

Background

- Finafloxacin is a novel pH activated, broad spectrum fluoroquinolone in development for infection indications in the hospital and critical care setting [1]
- Finafloxacin exhibits enhanced activity at low pH and under other environmental conditions associated with infection [1, 2]
- Finafloxacin exhibits bactericidal activity against forms of quiescent growth, thought to be relevant *in vivo* e.g. non-growing cells, biofilms and persisters [3]
- Other fluoroquinolones lose activity under such conditions. Consequently, finafloxacin exhibited superior activity in a series of infection models [4, 5]
- The activity of finafloxacin under infection relevant conditions and against infection relevant growth forms in combination with the high dosing potential predicted from its safety profile [6, 7], suggest finafloxacin will offer improved properties over currently marketed fluoroquinolones

Aim

Previous clinical studies have indicated that finafloxacin is well tolerated with few treatment-related AEs. As a part of the clinical development of finafloxacin, other PK studies are required to determine the effect of other variables on the PK profile of finafloxacin.

The primary objective of the study was as follows:

- To assess the PK profile of finafloxacin in healthy young and elderly volunteers.

The secondary objective of the study was as follows:

- To determine the safety and tolerability of finafloxacin in healthy young and elderly volunteers.

Methods

- All pertinent study documents were reviewed by the independently functioning Celerion Institutional Review Board (IRB) prior to study initiation. The IRB operations are in compliance with the U.S. Code of Federal Regulations (21 CFR Part 56) and International Conference on Harmonisation (ICH) guidelines. This study was conducted under the FDA investigational new drug number IND 106,676.
- This was a single-center, open-label, single-dose study in 40 healthy subjects. The subjects were assigned to 1 of 4 groups comprised of the following: 10 healthy young adult males, 10 healthy young adult females, 10 healthy elderly adult males, and 10 healthy elderly adult females.
- The subjects fasted for 10 hours and were administered 400 mg finafloxacin (as 2 x 200 mg tablets) administered with 240 mL of water.
- Blood samples were withdrawn at the following times, predose and at 30 minutes, 45 minutes, 1, 1.5, 2, 2.5, 3, 4, 6, 8, 12 and 24 hours postdose.
- Urine samples were collected from subjects over a 24-hour period postdose at the following intervals: baseline (predose) sample, 0 – 4 hours, 4 – 8 hours, 8 – 12 hours, and 12 – 24 hours.
- Finafloxacin PK parameters were summarized using descriptive statistics. The comparisons between the four age-gender groups administered 400 mg finafloxacin were assessed using an analysis of variance (ANOVA).
- The safety assessments included laboratory evaluations, physical examinations, AEs, standard 12-lead ECG parameters, and vital sign assessments

References

(1) Stubbings *et al.* 2010. *Antimicrobial Agents and Chemotherapy* In submission
(2) Kresken *et al.* 2008. 48th ICAAC / 46th IDSA Washington DC. Poster F1-2037.
(3) Goh *et al.* 2008. 48th ICAAC / 46th IDSA Washington DC. Poster F1-2042.
(4) Endermann *et al.* 2008. 48th ICAAC / 46th IDSA Washington DC. Poster F1-2044.
(5) Endermann *et al.* 2008. 48th ICAAC / 46th IDSA Washington DC. Poster F1-2045.
(6) Schmuck *et al.* 2008. 48th ICAAC / 46th IDSA Washington DC. Poster F1-2047.
(7) Patel *et* al. 2010. *Antimicrobial Agents and Chemotherapy* In submission.

Results

Summary of the Mean Pharmacokinetic Parameters for Plasma Finafloxacin in all Treatment Groups

Pharmacokinetic Parameters	Treatment Group Young Males (N)	Young Females (N)	Elderly Males (N)	Elderly Females (N)
Geometric Mean (Geometric CV%)				
C_{max} (ng/mL)	5160 (15.3) (10)	6440 (19.2) (10)	4630 (22.1) (10)	6190 (19.1) (10)
AUC_{0-t} (ng·h/mL)	16534 (35.4) (10)	18555 (23.5) (10)	19498 (21.6) (10)	22472 (19.7) (10)
AUC_{0-24} (ng·h/mL)	16534 (35.4) (10)	18554 (23.5) (10)	19497 (21.5) (10)	22463 (19.7) (10)
AUC_{0-inf} (ng·h/mL)	17240 (40.4) (8)	19942 (25.2) (8)	20250 (22.2) (10)	23477 (18.9) (10)
Arithmetic Mean ± SD				
$t_{1/2}$ (h)	7.64 ± 2.01 (8)	7.87 ± 1.64 (8)	6.06 ± 0.961 (10)	7.09 ± 1.39 (10)
CL/F (L/h/kg)	0.354 ± 0.123 (8)	0.319 ± 0.0690 (8)	0.247 ± 0.0511 (10)	0.264 ± 0.0571 (10)
Vz /F (L/kg)	4.01 ± 1.92 (8)	3.68 ± 1.31 (8)	2.16 ± 0.574 (10)	2.78 ± 1.11 (10)
AUC/AUC_{0-inf} (%)	95.6 ± 2.01 (8)	96.3 ± 1.47 (8)	96.3 ± 1.58 (10)	95.7 ± 1.57 (10)
Median (Minimum – Maximum)				
T_{max} (h)	0.999 (0.500, 1.50) (10)	0.996 (0.749, 1.50) (10)	0.877 (0.499, 2.50) (10)	0.876 (0.509, 1.50) (10)

Abbreviations: AUC; area under the plasma concentration-versus-time curve, CL/F; total body clearance calculated as: Dose / AUC_{0-inf}, C_{max}; maximum measured plasma concentration, $t_{1/2}$; apparent terminal elimination half-life, T_{max}; time of the maximum measured plasma concentration, Vz /F; apparent volume of distribution at the terminal phase.

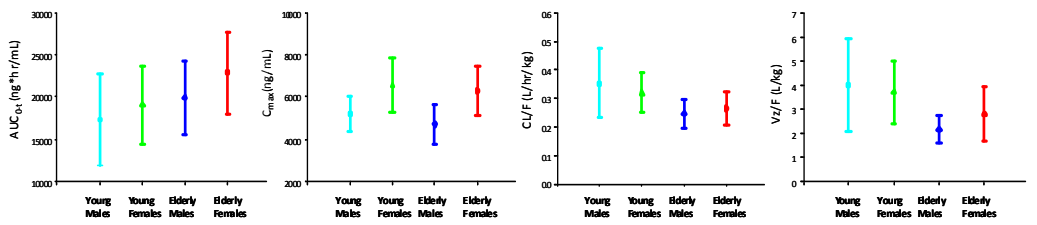

AUC0-t (ng*hr/mL)
Cmax (ng/mL)
CL/F (L/hr/kg)
Vz/F (L/kg)
Young Males
Young Females
Elderly Males
Elderly Females

Summary of the Mean Pharmacokinetic Parameters for Urine Finafloxacin in all Treatment Groups

Pharmacokinetic Parameters	Treatment Group Young Males (N)	Young Females (N)	Elderly Males (N)	Elderly Females (N)
Geometric Mean (Geometric CV%)				
$CumAe_{0-24}$ (mg)	132 (45.8) (10)	115 (23.4) (10)	84.3 (22.2) (10)	85.5 (28.5) (10)
R_{max} (mg/h)	18.2 (39.2) (10)	15.8 (27.7) (10)	9.68 (33.9) (10)	11.9 (26.2) (10)
Arithmetic Mean ± SD				
CumDose Excr. (%)	35.5 ± 12.9 (10)	29.5 ± 7.5 (10)	21.5 ± 4.8 (10)	22.2 ± 6.3 (10)
CLr (mL/h/kg)	120 ± 26.7 (10)	100 ± 21.2 (10)	54.9 ± 15.0 (10)	58.4 ± 9.6 (10)
Median (Minimum – Maximum)				
T_{max} (h)	2.02 (2.00, 2.03) (10)	2.01 (1.98, 2.02) (10)	2.06 (2.00, 2.08) (10)	2.03 (1.96, 2.05) (10)

Abbreviations: CLr; Renal clearance, CumAe0-24; Cumulative amount excreted in urine, CumDose Excr.; finafloxacin urinary excretion, R_{max}; maximum rate of urinary excretion, T_{max}; time of the maximum measured urine concentration,

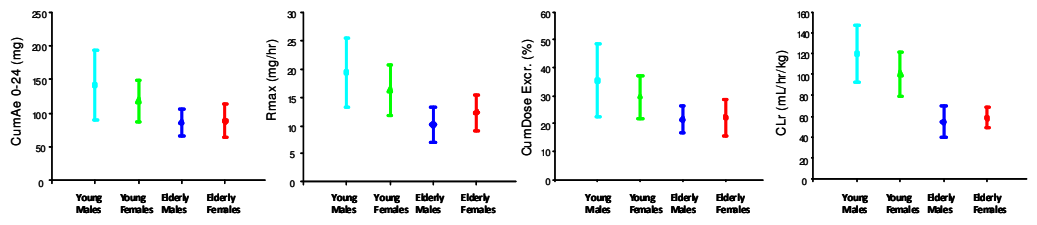

CumAe 0-24 (mg)
Rmax (mg/hr)
CumDose Excr. (%)
CLr (mL/hr/kg)
Young Males
Young Females
Elderly Males
Elderly Females

Summary of Adverse Event (AE) Incidence by Treatment Group

- 22 / 23 AEs were classified by the principal investigator (PI) as mild in intensity and 1 /23 as moderate
- The most common AEs were headache and nausea
- There were no significant findings in clinical laboratory, vital signs, ECGs, and physical examinations.

Treatment Group	Subject Incidence (%)	Number of AEs	Number of Treatment-Related* AEs
Young Males (n=10)	4 (40%)	6	6
Young Females (n=10)	6 (60%)	11	6
Elderly Males (n=10)	2 (20%)	2	2
Elderly Females (n=10)	3 (30%)	4	2
Total (n=40)	15 (38%)	23	16

*= At least possibly related to study treatment

Conclusions

- The average systemic availability ($AUC_{0-inf.}$) of finafloxacin was approximately 18% greater in elderly versus young subjects (not statistically significant). Mean systemic exposure was similar in males and females (<16% difference).
- The mean peak exposure (C_{max}) of FIN was similar in elderly (4630 - 6190 ng/mL) and young (5160 - 6440) subjects, but slightly higher in females versus males.
- Urinary excretion of finafloxacin (based on $CumAe_{0-24}$ and R_{max}) was significantly lower (by 42% – 54%) in elderly compared to young subjects and was similar between genders.
- There were no statistically significant age or gender effects on FIN PK except on urinary excretion (age) and peak exposure (gender).
- Overall, there were no major safety concerns found in the vital sign, safety laboratory, ECG, AE, or physical examination assessments associated with the administration of finafloxacin in young and elderly healthy male and female subjects.
- The administration of a single oral dose of 400 mg finafloxacin was safe and well tolerated in the young and elderly male and female subjects in this study.